Plasmodium falciparum parasite dynamics determined by qPCR after Controlled Human Malaria Infection in Semi-Immunes from Gabon.

Honkpehedji J[1], Dejon Agobe JC[1], Zinsou J[1], Mengue J[2], Bouyoukou Hounkpatin A[2] , Held J, Adegnika AA[1,2], Richie T[3], Mordmüller B[1,2], Kremsner PG[1,2], Hoffman SL[3], Lell B[1,2].

[1] Centre de Recherches Médicales de Lambaréné, Gabon,
[2] Institute for Tropical Medicine, University of Tübingen, Tübingen, Germany,
[3] Sanaria Inc., Rockville, MD, United States.

BACKGROUND

Sanaria has developed aseptic, purified, cryopreserved infectious *Plasmodium falciparum* (Pf) sporozoites (SPZ) called Sanaria® PfSPZ Challenge as a tool for Controlled Human Malaria Infections (CHMI) to study protective efficacy of antimalarial vaccines, and to allow refinement of the method of administration of the highly protective PfSPZ Vaccine. A critical component of the CHMI studies with PfSPZ Challenge is the diagnosis of malaria parasites in the blood. Characterising the effect of natural acquired immunity and sickle cell anaemia on the pattern of *Pf* parasitemia may be useful in understanding the pathophysiological mechanisms of protection against malaria. CHMI by direct venous inoculation of PfSPZ Challenge is a new tool which can be used to investigate the pathophysiology of malaria.

METHODS

The study was performed in Lambaréné, Gabon, one of seven African partners in the EDCTP-funded CHMI platform. Adults aged 18-35 from three groups : 5 non-immune (NI), 11 semi-immunes with hemoglobin AA (IA), and 9 semi-immunes with hemoglobin AS (IS) received 3200 sporozoites after a curative treatment cure with clindamycin. Capillary blood samples were taken daily up to Day 28 to determine parasitemia by real time quantitative polymerase chain reaction (RT-qPCR) as described by Hermsen CC et al (2001) . Treatment was administered for a malaria episode or at Day 28, whichever came first.

RESULTS

Parasitemia was detected in 5 (100%) subjects in the NI group, 9 (82%) in the IA group and 7 (78%) in the IS group. All volunteers in the NI group showed similar patterns with parasitemia starting around Day 8 and rising quickly. Patterns for parasitemia in the immune groups (IA and IS) were highly heterogeneous. Although time-points of initial parasitemia and duration of parasitemia were varied, all semi-immunes managed to control parasitemia for at least several days. There were no discernible differences in patterns between the IS and IA group, including the area under curve of parasitemia over time.

Fig. Parasitemia detected by PCR

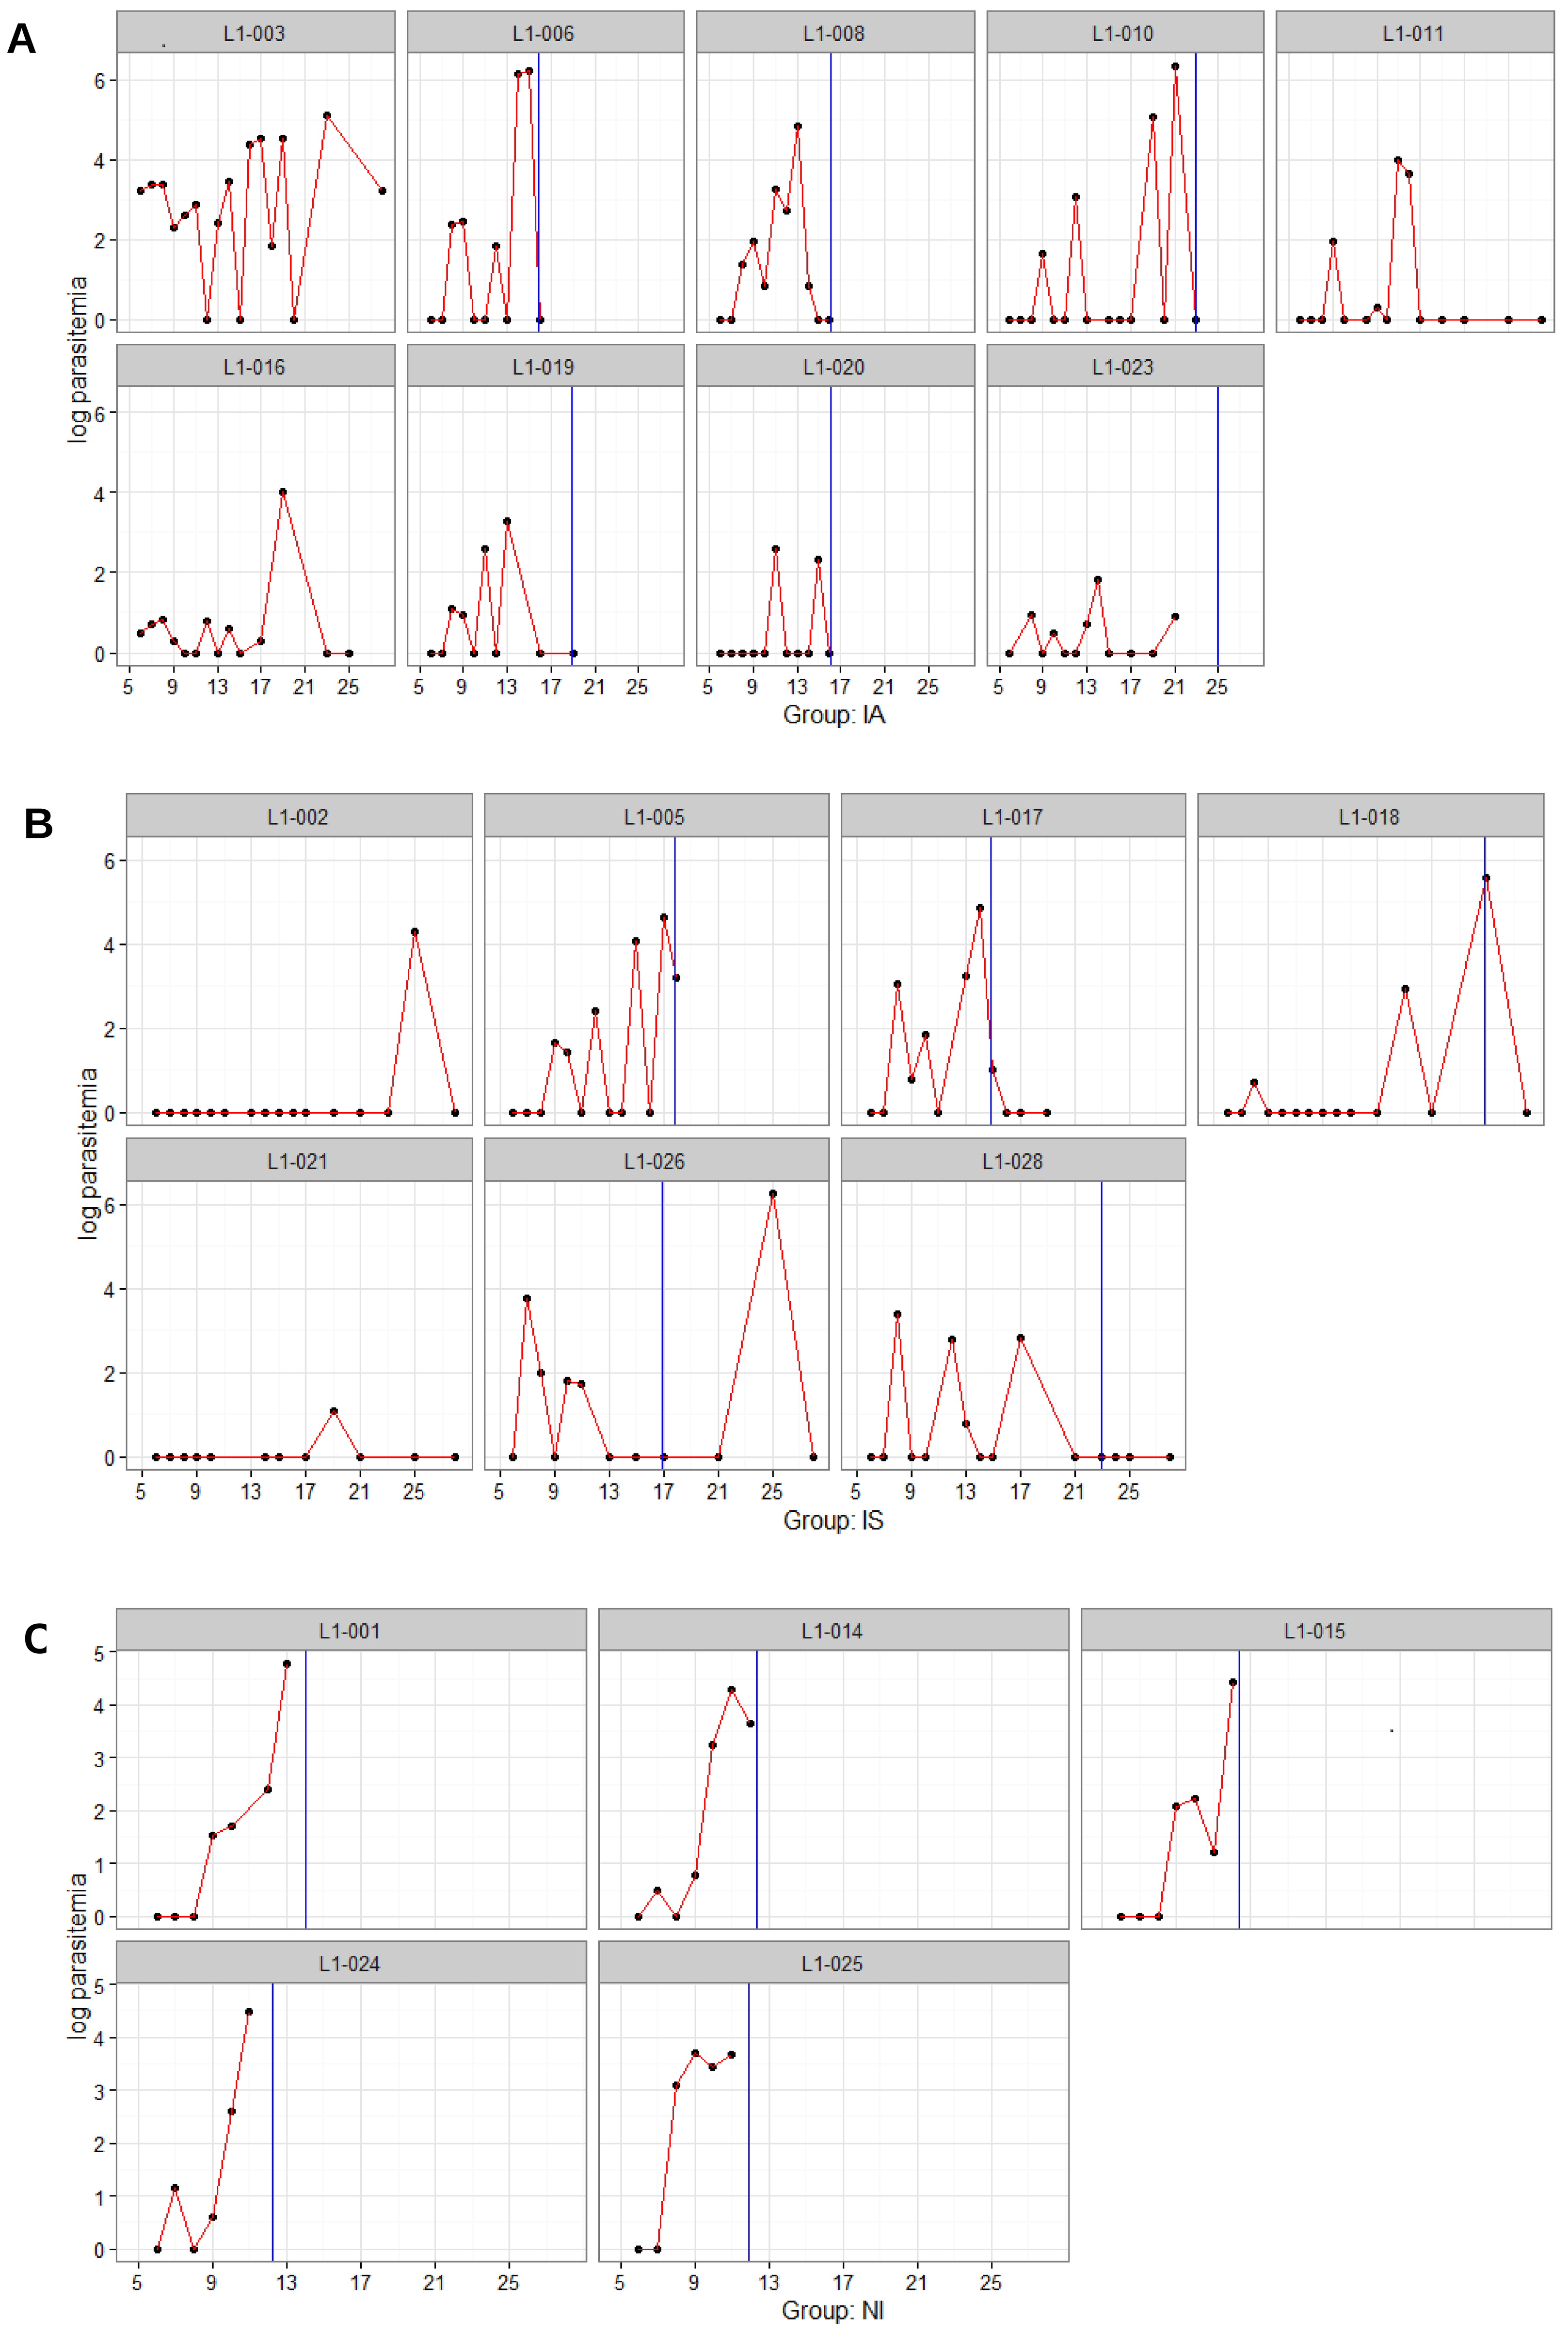
A
L1-003
L1-006
L1-008
L1-010
L1-011
L1-016
L1-019
L1-020
L1-023
log parasitemia
Group: IA
B
L1-002
L1-005
L1-017
L1-018
L1-021
L1-026
L1-028
log parasitemia
Group: IS
C
L1-001
L1-014
L1-015
L1-024
L1-025
log parasitemia
Group: NI

The vertical blue line shows the time points of onset of symptoms

CONCLUSION

No parasitemia was detected in 20% of the semi-immunes, likely due to liver stage immunity. The highly variable patterns of parasitemia do not allow us to discern immune mechanisms against blood stages. Hemoglobin AS had no visible effect on parasite dynamics at the low parasitemia encountered.

REFERENCES

1- Ishizuka AS et al. Protection against malaria at 1 year and immune correlates following PfSPZ vaccination. Nat Med. 9. Mai 2016
2- Hermsen CC et al. Detection of *Plasmodium falciparum* malaria parasites in vivo by real-time quantitative PCR. Mol Biochem Parasitol. 2001;118:247–51. doi: 10.1016/S0166-6851(01)00379-6.